Switching botulinum toxin formulations from onabotulinumtoxinA (Botox®) to incobotulinumtoxinA (Xeomin®): experience from a spasticity outpatient clinic

Prabal K. Datta, Adrian Robertson

Mid Yorkshire Hospitals NHS Trust, Wakefield, UK

Background

- Spasticity related to neurologic disease is relatively common and can be troublesome to affected individuals.[1]
- Botulinum toxin type A (BoNT-A) injections are a recommended treatment option for limb spasticity due to various neurologic conditions.[2–4]
- IncobotulinumtoxinA (Xeomin®, Merz Pharmaceuticals GmbH) treatment resulted in significant improvements in spasticity-associated disability and muscle tone, and was well tolerated in previous randomized, placebo-controlled clinical trials with open-label extension periods.[5–7]
- In 2009, the Mid Yorkshire Hospitals NHS Trust made the decision to switch from onabotulinumtoxinA (Botox®, Allergan Inc.) to incobotulinumtoxinA for the management of spasticity related to neurologic conditions.
- In order to assess the clinical impact of this switch in BoNT-A treatment, a retrospective case-file review was performed to examine dose requirements, treatment intervals, and tolerability of onabotulinumtoxinA and incobotulinumtoxinA.

Methods

Study design and setting

- Retrospective case-file review of consecutive patients in an outpatient spasticity-management clinic.

Inclusion and exclusion criteria

- Patients with spasticity related to any neurologic condition, who had previously been treated with onabotulinumtoxinA and were switched to incobotulinumtoxinA (**Table 1**).

Table 1. Inclusion and exclusion criteria

Inclusion criteria	Exclusion criteria
Any patient switched from onabotulinumtoxinA to incobotulinumtoxinA within the last 7 years	Any patient not switched from onabotulinumtoxinA to incobotulinumtoxinA within the last 7 years
Age ⩾18 years	Age <18 years
Diagnosis of spasticity in upper or lower limbs	Insertion of intrathecal baclofen pump during study period Initiation of oral antispasmodic medication during study period Limb fractures during the study period Limb surgery for spasticity and/or contracture during the study period

Interventions

- BoNT-A was injected into the affected muscles of the upper and/or lower limb.
- For each treatment, dosing, injection sites, and treatment intervals were adjusted based on clinical need and previous treatment outcomes.
- Switching from onabotulinumtoxinA to incobotulinumtoxinA was generally initiated at a unit dose ratio of 1:1. Both products were reconstituted to the same volume. Electromyography, electrostimulation or ultrasound were occasionally used to guide injections. No change in practice took place during the study period.

Main outcome measures

- Patient records were used to document treatment intervals, doses, muscles treated, injection technique, and adverse reactions.

Results

Study population and baseline characteristics

- Records from 254 consecutive patient records were reviewed; 93 patients fulfilled the inclusion criteria. A total of 161 patient records were excluded from the review (**Table 2**).
- Patients included were 16–82 years of age (mean 46.5 years) at the start of treatment, and 59% were male (**Table 3**).
- Spasticity was mainly due to stroke (40.9%), cerebral palsy (25.8%) or multiple sclerosis (18.3%; **Table 3**).
- Patients had been treated with onabotulinumtoxinA for 3–55 months (mean 16 months) before receiving incobotulinumtoxinA for 7–73 months (mean 39 months) (**Table 3**).

Dosing interval and dose requirements

- The mean treatment intervals for onabotulinumtoxinA and incobotulinumtoxinA were similar (153.7 days and 155.4 days, respectively; **Figure 1**).
- The mean dose per limb per visit for onabotulinumtoxinA and incobotulinumtoxinA were also similar (144.8 U and 145.0 U, respectively; **Figure 2**).

Safety

- No adverse reactions occurred with either BoNT-A formulation.

Table 2. Reasons for exclusion

Reason for exclusion	Number of patients[a]
Treatment commenced after switch	90
Non-switch patients	52
Dystonia	4
Hypersalivation	2
No historic record available	2
Non-neurology	1
Double counted	9
Incorrect patient	1
Total exclusions	**161**

[a]Patients could have been excluded for more than one reason.

Table 3. Baseline characteristics

Baseline characteristic	Patients N=93
Mean age at start of treatment, years (range)	46.5 (16–82)
Male gender, n (%)	55 (59.1)
Neurologic condition, n (%)	
Stroke	38 (40.9)
Traumatic brain injury	7 (7.5)
Multiple sclerosis	17 (18.3)
Encephalitis	1 (1.1)
Hereditary spastic paraparesis	3 (3.2)
Cerebral palsy	24 (25.8)
Spinal cord injury	1 (1.1)
Other	2 (2.2)
Limbs treated, n	
Total limbs treated	127
Right leg	37
Left leg	36
Right arm	38
Left arm	16
Mean duration of condition, months (range)	237 (26–720)
Mean duration of BoNT-A treatment, months (range)	
Total	78 (13–240)
OnabotulinumtoxinA	16 (3–55)
IncobotulinumtoxinA	39 (7–73)

BoNT-A, botulinum neurotoxin type A.

Figure 1. Treatment intervals with onabotulinumtoxinA and incobotulinumtoxinA

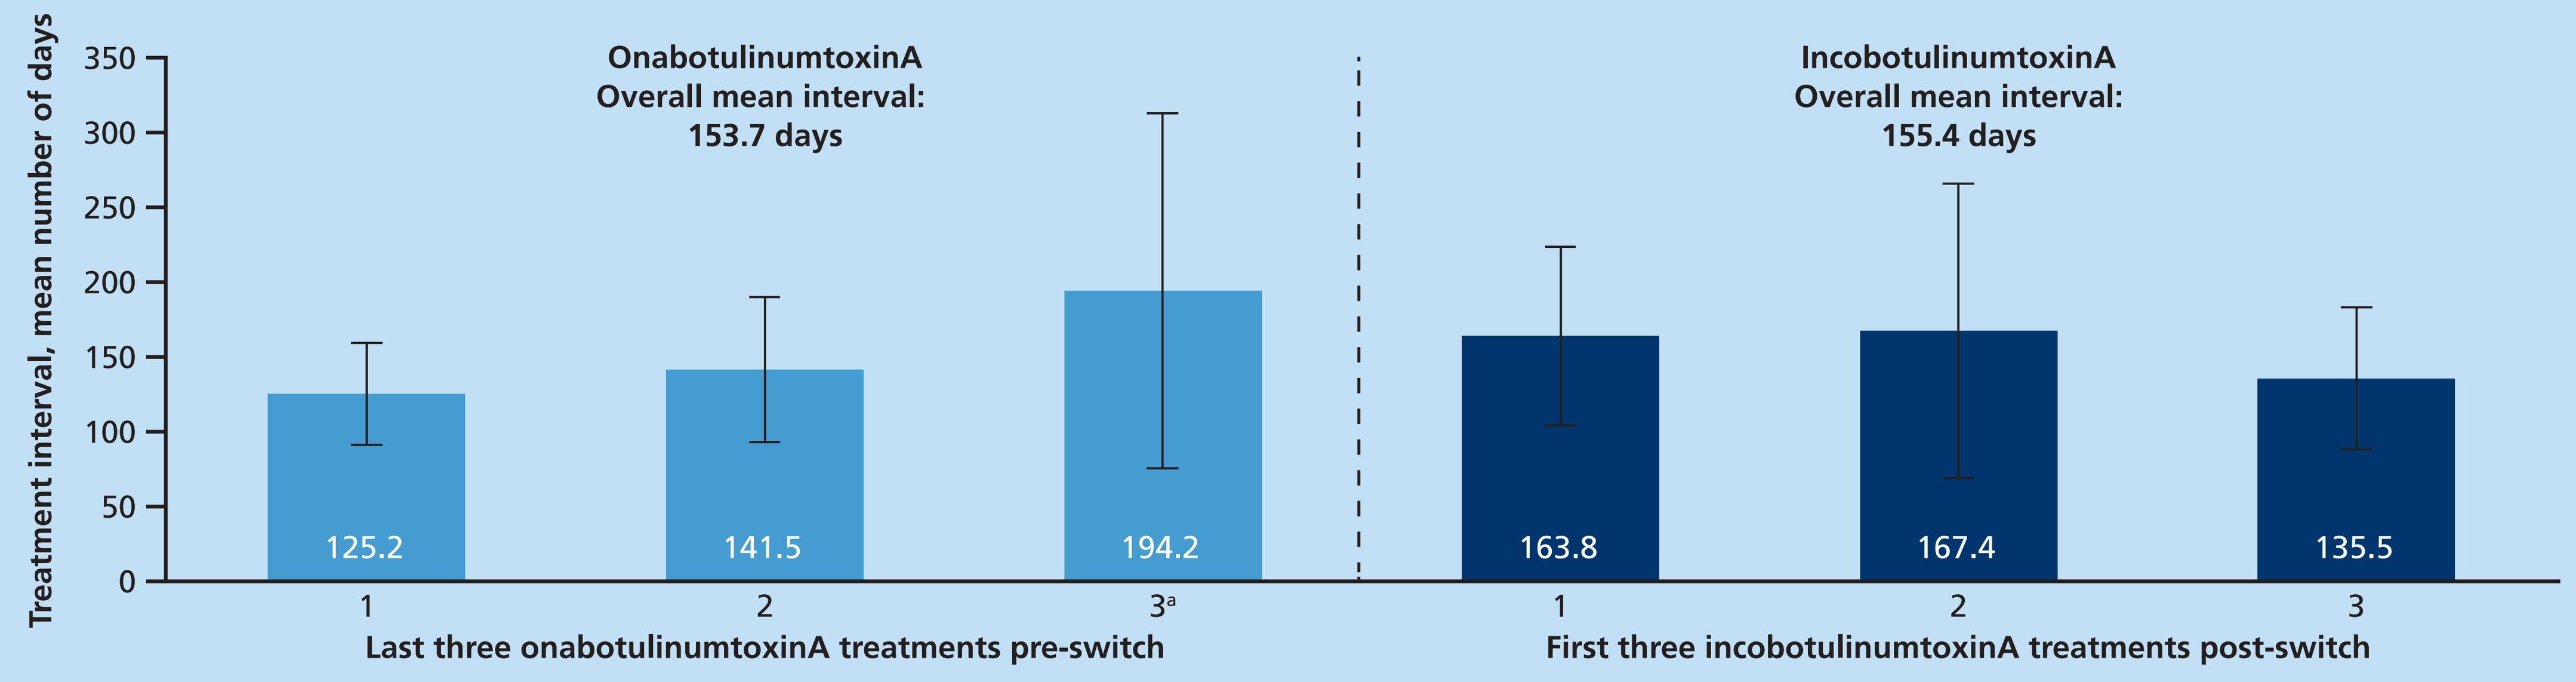
Treatment interval, mean number of days
350
300
250
200
150
100
50
0
OnabotulinumtoxinA
Overall mean interval:
153.7 days
IncobotulinumtoxinA
Overall mean interval:
155.4 days
125.2
141.5
194.2
163.8
167.4
135.5
1
2
3a
1
2
3
Last three onabotulinumtoxinA treatments pre-switch
First three incobotulinumtoxinA treatments post-switch

[a]Interval between the last onabotulinumtoxinA treatment and the first incobotulinumtoxinA treatment.
Error bars show the standard deviation.

Figure 2. Mean dose per limb per visit with onabotulinumtoxinA and incobotulinumtoxinA

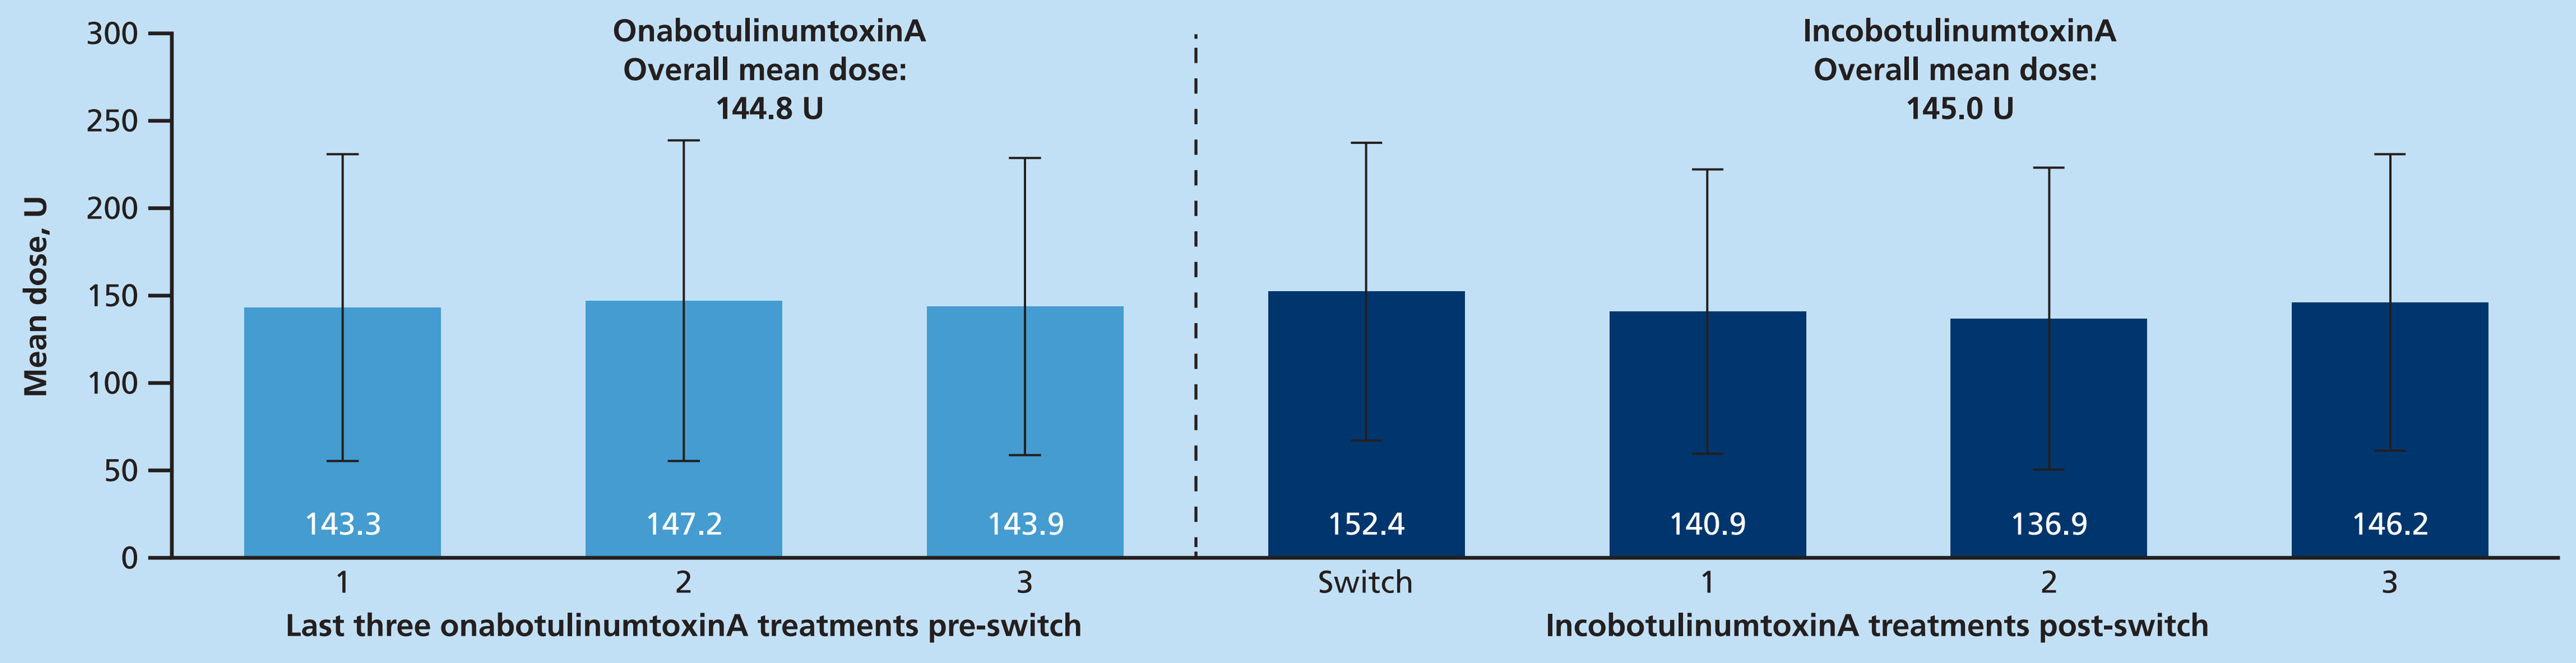
Mean dose, U
300
250
200
150
100
50
0
OnabotulinumtoxinA
Overall mean dose:
144.8 U
IncobotulinumtoxinA
Overall mean dose:
145.0 U
143.3
147.2
143.9
152.4
140.9
136.9
146.2
1
2
3
Switch
1
2
3
Last three onabotulinumtoxinA treatments pre-switch
IncobotulinumtoxinA treatments post-switch

Error bars show the standard deviation.

Conclusions

- Switching of onabotulinumtoxinA to incobotulinumtoxinA in a 1:1 ratio did not lead to any changes in the mean treatment interval or dose per visit. As these measures have previously been used as a proxy measure of the efficacy of BoNT-A treatment,[8] these results suggest that both onabotulinumtoxinA and incobotulinumtoxinA have a similar efficacy profile.
- No adverse reactions were observed with either onabotulinumtoxinA or incobotulinumtoxinA, indicating that both had similar safety and tolerability profiles.
- Overall, results indicate that onabotulinumtoxinA and incobotulinumtoxinA can be switched at a 1:1 unit dose ratio in clinical practice.

References

1. Zorowitz RD et al. Medscape 2008. Available at http://www.medscape.org/viewarticle/576698 [accessed 24 July 2015].
2. Esquenazi A et al. Toxicon 2013; 67: 115–128.
3. Wissel J et al. J Rehabil Med 2009; 41: 13–25.
4. Simpson DM et al. Neurology 2016; 86: 1818-1826.
5. Kaňovský P et al. Clin Neuropharmacol 2009; 32: 259–265.
6. Kaňovský P et al. J Rehabil Med 2011; 43: 486–492.
7. Elovic EP et al. Muscle & Nerve 2016; 53: 415-421.
8. Dressler D. Eur J Neurol 2009; 16 (Suppl 2): 2–5.

Acknowledgements

The authors have received a research grant from Merz Pharmaceuticals GmbH, Frankfurt am Main, Germany. Editorial support was provided by Mike Lappin, PhD, on behalf of Complete Medical Communications and funded by Merz Pharmaceuticals GmbH.

Presented at TOXINS 2017, Madrid, Spain, 18–21 January 2017

MRZ82220d-Dattar.indd 1
11/01/2017 14:48